ISPPD 2020

www.can-r.ca

PCV15 and PPSV23 Coverage of Invasive and Respiratory Tract Isolates of *Streptococcus pneumoniae*: CANWARD 2007-2018

A.R. GOLDEN[1], H.J. ADAM[1,2], M. BAXTER[1], I. MARTIN[3], W. DEMCZUK[3], M.R. MULVEY[1,3], J.A.KARLOWSKY[1,2], G.G. ZHANEL[1], and the CANADIAN ANTIMICROBIAL RESISTANCE ALLIANCE (CARA)

[1]University of Manitoba, [2]Shared Health and [3]National Microbiology Laboratory, Winnipeg, Canada

Dr. Alyssa Golden
Microbiology, Health Sciences Centre
MS673-820 Sherbrook St.
Winnipeg, MB R3A 1R9
Email: alyssa-golden@hotmail.com

Introduction

Streptococcus pneumoniae is a leading cause of both invasive and respiratory infections, and a significant source of morbidity and mortality worldwide (1). Numerous vaccines are available to help prevent pneumococcal disease caused by common serotypes. The 23-valent polysaccharide vaccine (PPSV23, including serotypes 1, 2, 3, 4, 5, 6B, 7F, 8, 9N, 9V, 10A, 11A, 12F, 14, 15B, 17F, 18C, 19A, 19F, 20, 22F, 23F and 33F) has been in use in Canada for years, particularly in adults over the age of 65 (2). Pneumococcal conjugate vaccines were introduced in Canada in the 2000s and are included as part of the routine infant immunization schedule (3). The currently recommended conjugate vaccine is PCV13 (serotypes 1, 3, 4, 5, 6A, 6B, 7F, 9V, 14, 18C, 19A, 19F, and 23F), however an expanded formulation (PCV15, including PCV13 serotypes plus 22F and 33F) is currently undergoing clinical trials (3,4).

The objective of this study was to compare the proportion of invasive and respiratory tract isolates of *S. pneumoniae*, including multidrug- and extensive-drug resistant (MDR, XDR) strains, that demonstrated PCV15 and PPSV23 serotypes in Canada from 2007 to 2018.

Materials and Methods

Bacterial Isolates

As part of the ongoing CANWARD study, Canadian hospital laboratories were asked to submit clinical isolates (consecutive, one per patient, per infection site) that were clinically significant. Isolates were shipped to the coordinating laboratory (Health Sciences Centre, Winnipeg, Canada) where they were subcultured onto appropriate media and stocked in skim milk at -80° C. The CANWARD study collected 2821 *S. pneumoniae* isolates from 2007-2018 (986 invasive, 1835 respiratory).

Antimicrobial Susceptibility Testing and Serotyping

Following 2 subcultures from frozen stock, *in vitro* antimicrobial susceptibility testing was performed using the Clinical and Laboratory Standards Institute (CLSI) broth microdilution method (5). MIC interpretive criteria were defined according to CLSI breakpoints (6). MDR and XDR were defined as resistance to ≥3 and ≥5 antimicrobial classes, respectively. Serotyping was performed using the Quellung reaction and commercial antisera.

Results

Table 1. Proportion of blood and respiratory *S. pneumoniae* demonstrating a PCV15 or PPSV23 serotype, by age group

Age Group (years)	Vaccine Coverage (%)					
	PCV15			PPSV23		
	Blood	Respiratory	*P*-value [a]	Blood	Respiratory	*P*-value [a]
0-<2	51.6	40.3	NS	71.9	54.8	NS
2-<6	70.8	34.4	<0.0001	81.5	47.9	<0.0001
6-<18	72.9	40.9	0.0006	81.3	56.8	0.0046
18-<50	52.4	39.3	0.0011	82.7	58.5	<0.0001
50-<65	54.0	40.4	0.0005	75.0	55.9	<0.0001
≥65	53.7	39.3	<0.0001	72.5	53.6	<0.0001

[a] *P*-value for the proportion of blood isolates vs. the proportion of respiratory isolates for the given vaccine and demographic category. NS, not significant.

Table 2. Proportion of blood and respiratory *S. pneumoniae* demonstrating a PCV15 or PPSV23 serotype, by region

Canadian Region [a]	Vaccine Coverage (%)					
	PCV15			PPSV23		
	Blood	Respiratory	*P*-value [b]	Blood	Respiratory	*P*-value [b]
Western	53.2	36.7	<0.0001	80.3	55.6	<0.0001
Central	57.9	41.8	<0.0001	75.3	54.6	<0.0001
Eastern	50.0	39.6	NS	67.9	56.4	NS

[a] Western Canada includes British Columbia, Alberta, Saskatchewan and Manitoba; Central Canada includes Ontario and Quebec; Eastern Canada includes New Brunswick and Nova Scotia. [b] *P*-value for the proportion of blood isolates vs. the proportion of respiratory isolates for the given vaccine and demographic category. NS, not significant.

Table 3. Proportion of blood and respiratory *S. pneumoniae* demonstrating a PCV15 or PPSV23 serotype, by gender

Gender	Vaccine Coverage (%)					
	PCV15			PPSV23		
	Blood	Respiratory	*P*-value [a]	Blood	Respiratory	*P*-value [a]
Female	55.5	40.8	<0.0001	75.5	55.7	<0.0001
Male	55.3	38.6	<0.0001	77.6	55.1	<0.0001

[a] *P*-value for the proportion of blood isolates vs. the proportion of respiratory isolates for the given vaccine and demographic category. NS, not significant.

Table 4. Proportion of blood and respiratory *S. pneumoniae* demonstrating a PCV15 or PPSV23 serotype, by antimicrobial resistance phenotype

Resistance Phenotype	Vaccine Coverage (%)					
	PCV15			PPSV23		
	Blood	Respiratory	*P*-value [a]	Blood	Respiratory	*P*-value [a]
Clarithromycin-R	60.5	49.3	0.0246	75.7	57.8	<0.0001
Doxycycline-R	51.3	45.1	NS	60.3	50.4	NS
Penicillin-R [b]	89.7	76.9	NS	93.1	79.1	NS
TMP/SMX-R	74.2	63.0	NS	83.9	64.7	0.0058
MDR	64.4	54.3	NS	71.1	56.1	NS
XDR	92.9	88.9	NS	92.9	88.9	NS

[a] *P*-value for the proportion of blood vs. the proportion of respiratory for the given vaccine and antimicrobial resistance phenotype.
[b] oral penicillin V breakpoints (resistant MIC ≥2 mg/L). NS, not significant.

Conclusions

1. Overall, the proportion of blood isolates demonstrating a PCV15/PPSV23 serotype was significantly higher than respiratory strains (55.4/76.7% vs 39.5/55.3%, *P*<0.0001).
2. By age group, with the exception of the <2-year category, the proportion of blood isolates demonstrating a PCV15/PPSV23 serotype was significantly higher than respiratory strains (*P*≤0.0046).
3. The proportion of blood isolates demonstrating a PCV15/PPSV23 serotype was significantly higher by gender (*P*<0.0001) and region (*P*≤0.0002), with the exception of Eastern Canada.
4. There was no significant difference in the proportion of doxycycline- or penicillin-resistant blood and respiratory isolates demonstrating PCV15/PPSV23 serotypes. The proportion of clarithromycin-resistant isolates from blood demonstrating a PCV15/PPSV23 serotype was significantly higher than respiratory strains (*P*≤0.0246). Similarly, the proportion of trimethoprim/sulfamethoxazole-resistant isolates from blood demonstrating a PPSV23 serotype was significantly higher than respiratory strains (0.00058), but not for PCV15 serotypes.
5. There was no significant difference in the proportion of MDR blood (64.4/71.1%) and respiratory (54.3/56.1%) isolates representing PCV15/PPSV23 serotypes, respectively.

Acknowledgements

The authors thank Merck for their support of this study. The authors would also like to thank the Streptococcus and STI Group at the National Microbiology Laboratory, and the participating CANWARD centres, investigators and laboratory site staff for their continued support:

Vancouver Hospital, Vancouver; University of Alberta Hospital, Edmonton; Royal University Hospital, Saskatoon; Health Sciences Centre, Winnipeg; London Health Sciences Centre, London; Hamilton Health Sciences Corporation, Hamilton; St. Joseph's Hospital, Hamilton; University Health Network & Mount Sinai Hospital, Toronto; St. Michael's Hospital, Toronto; Children's Hospital of Eastern Ontario, Ottawa; The Ottawa Hospital, Ottawa; Royal Victoria Hospital, Montreal; Montreal General Hospital, Montreal; Sir Mortimer B. Davis Jewish General Hospital, Montreal; Hôpital de la Cité-de-la-Santé, Laval; Hôpital Maisonneuve-Rosemont, Montreal; CHRTR Pavilion Ste. Marie, Trois-Rivières; Centre Hospitalier Universitaire de Sherbrooke, Sherbrooke; L'Hotel-Dieu de Quebec, Quebec City; South East Regional Health Authority, Moncton; Queen Elizabeth II Health Sciences Centre, Halifax.

References

1. Lynch JP and Zhanel GG. *Curr Opin Pulm Med.* 2010; 16: 217-25.
2. Desai S, et al. *Can Commun Dis Rep*. 2010; 36:1-21.
3. Quach C, et al. *Can Commun Dis Rep.* 2016; 42: 260-62.
4. Stacey HL et al. *Hum Vaccine Immunother*. 2019; 15: 530-39.
5. CLSI. M07, 11th Edition. Wayne, PA. 2018.
6. CLSI. M100, 30th Edition. Wayne, PA. 2020.