110P ESMO 2022

Characteristics and survival outcomes of patients with *RET* fusion-positive (*RET*-fp) solid tumours receiving non-RET inhibitor therapy in a real-world setting

Pilar Garrido[1], Salvatore Siena[2], Matthew Taylor[3], Walter Bordogna[4], Otto Fajardo[4], Christos Nikolaidis[4]

BACKGROUND

- *RET* gene fusions are oncogenic drivers in multiple tumour types.[1,2]
- Current standards of care for patients with *RET*-fp solid tumours have limited efficacy and/or significant off-target toxicity;[2,3] there is a need for efficacious precision therapies that selectively target *RET* alterations.
- The genomic landscape and natural history of patients with *RET*-fp solid tumours is unknown.
- Using real-world data, we described the clinical characteristics and survival outcomes of patients with *RET*-fp metastatic solid tumours who received non-selective RET inhibitor therapy, and assessed the prognostic value of *RET* fusions in solid tumours.

METHODS

Study design

- Clinical characteristics and survival outcomes for RET inhibitor-naïve patients with metastatic solid tumours were collected from the nationwide (US-based) de-identified Flatiron Health/Foundation Medicine clinico-genomic database (FH-FMI CGDB; version April 2022)
 - The de-identified data originated from ~280 cancer clinics in the US (~800 care sites); retrospective longitudinal clinical data were derived from electronic health records, comprising patient-level structured and unstructured data, curated via technology-enabled abstraction, and were linked to genomic data derived from Foundation Medicine comprehensive genomic profiling (CGP) tests in the FH-FMI CGDB by de-identified, deterministic matching.[4]
- Eligible patients had:
 - ≥1 documented clinical visit in the Flatiron Health network between 1 January 2011 and 31 March 2022, and underwent ≥1 DNA Foundation Medicine CGP test prior to 1 April 2022
 - A visit gap ≤90 days after initial diagnosis.
- Patients with a diagnosis of *RET*-fp non-small cell lung cancer were excluded.
- *RET* positivity was defined by the presence of a fusion with a predicted known/likely functional status.
- A *RET* wild-type (*RET*-WT) status was determined when the CGP test was unable to detect qualifying *RET* fusions.

Matching of *RET*-WT and *RET*-fp patients

- Mahalanobis distance matching was used to match *RET*-WT with *RET*-fp patients in a 4:1 ratio, using two models:
 - In both models, patients were matched using preselected covariates: age, gender, race, tumour type, practice type, Eastern Cooperative Oncology Group (ECOG) performance status, year of CGP, time from initial diagnosis to CGP report date and number of treatment lines prior to the CGP report date
 - In model 1, the CGP report date was used as the index date; in model 2, the date of metastatic diagnosis (after adjustment for immortal time bias) was used as the index date.

Objectives

- The primary objective was to evaluate overall survival (OS) in the *RET*-fp and covariate-matched *RET*-WT cohorts.
- Secondary objectives included descriptions of patient characteristics, treatment patterns and genomic alterations
 - Tumour mutational burden (TMB), microsatellite instability (MSI) and oncogenic alterations in *ALK*, *BRAF*, *ERBB2*, *EGFR*, *NTRK*, *ROS1*, *MET* and *KRAS* were assessed.

RESULTS

Patient characteristics

- Overall, there were 7,220 eligible patients with *RET*-WT solid tumours.
- The *RET*-fp cohort comprised 26 patients and the matched *RET*-WT cohort comprised 104 patients (**Table 1**).

Table 1. Baseline characteristics

Characteristic	*RET*-fp (N=26)	Matched *RET*-WT (N=104)	Non-matched *RET*-WT (N=7,116)
Mean age, years (SD)	65.3 (10.3)	61.9 (12.0)	64.5 (10.1)
Sex,* n (%)			
Female	11 (42.3)	44 (42.3)	4,033 (56.7)
Male	15 (57.7)	60 (57.7)	3,082 (43.3)
Race, n (%)			
Asian / Black or African American	0 / 1 (3.8)	0 / 4 (3.8)	173 (2.4) / 615 (8.6)
Hispanic or Latino / White	0 / 21 (80.8)	0 / 91 (87.5)	15 (0.2) / 4,597 (64.6)
Other / Missing	3 (11.5) / 1 (3.8)	9 (8.7) / 0	1,120 (15.7) / 596 (8.4)
ECOG PS†, n (%)			
0	7 (26.9)	26 (25.0)	1,656 (23.3)
1	7 (26.9)	34 (32.7)	2,339 (32.9)
≥2	1 (3.8)	4 (3.8)	869 (12.2)
Missing	11 (38.5)	40 (38.5)	2,252 (31.6)
Prior lines of therapy, n (%)			
0	2 (7.7)	8 (7.7)	807 (11.3)
1	6 (32.1)	24 (32.1)	2,580 (26.3)
≥2	7 (26.9)	28 (26.9)	1,932 (27.2)
Missing	11 (42.3)	44 (42.3)	1,797 (25.3)
Practice type, n (%)			
Academic	4 (15.4)	15 (14.4)	999 (14.0)
Community	22 (84.6)	89 (85.6)	6,117 (86.0)
Mean time from initial diagnosis to CGP report date, months (SD)	8.1 (10.7)	7.9 (8.6)	14.5 (22.9)

*Data missing for one patient in the non-matched *RET*-WT cohort. †Closest ECOG PS 30 days prior to 7 days after CGP report date. CGP, comprehensive genomic profiling; ECOG PS, Eastern Cooperative Oncology Group performance status; *RET*-fp, *RET* fusion-positive; *RET*-WT, *RET* wild type.

- In total, nine distinct *RET*-fp tumour types were identified; the most frequent were colorectal (34.6%), pancreatic (15.4%) and thyroid (15.4%) cancers (**Figure 1**).
- Of nine different *RET* fusion partners detected, the most common were *NCOA4* (46.2%), *CCDC6* (23.1%) and *ERC1* (7.7%).

Figure 1. Tumour types in the *RET*-fp cohort (N=26)

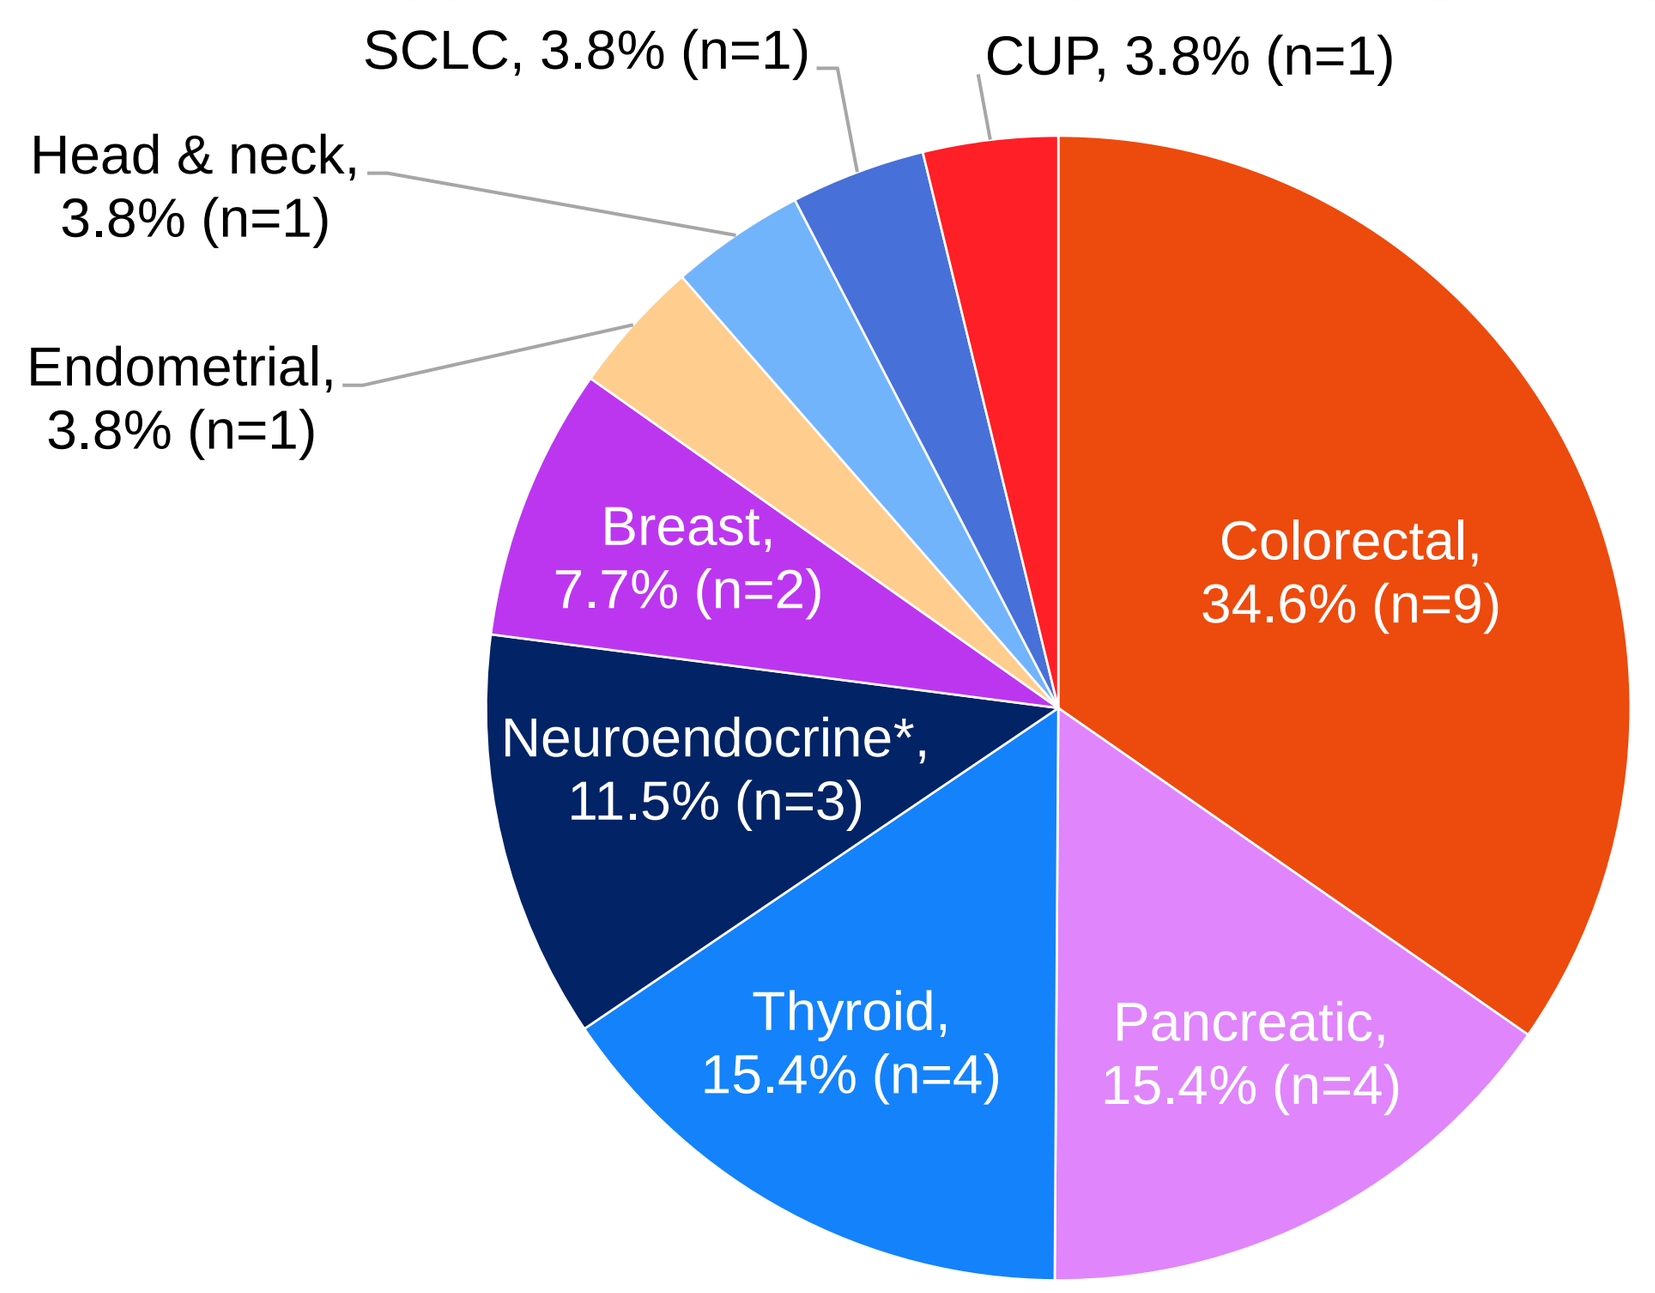
SCLC, 3.8% (n=1)
CUP, 3.8% (n=1)
Head & neck, 3.8% (n=1)
Endometrial, 3.8% (n=1)
Breast, 7.7% (n=2)
Neuroendocrine*, 11.5% (n=3)
Thyroid, 15.4% (n=4)
Pancreatic, 15.4% (n=4)
Colorectal, 34.6% (n=9)

Patients with *RET*-fp (NSCLC excluded). *Neuroendocrine tumours included one gastrointestinal tumour and two unspecified anatomical locations. CUP, cancer of unknown primary; NSCLC, non-small cell lung cancer; *RET*-fp, *RET* fusion-positive; SCLC, small cell lung cancer.

TMB and MSI

- In the *RET*-fp cohort, 65.4% of patients had a low TMB status (7.7% were TMB-high) and 50.0% had a low MSI status (3.8% were MSI-high) (**Table 2**)
 - One patient in the *RET*-fp cohort had high MSI and an *ERBB2* amplification
 - No other assessed oncogenic co-alterations were identified in this cohort.
- Most patients in the matched *RET*-WT cohort also had low levels of TMB (84.6%) and MSI (82.7%) (**Table 2**)
 - Patients in this cohort had an equal rate of *ERBB2* (3.8%), and a higher rate of *BRAF* (5.8%) and *KRAS* (37.5%) alterations than patients in the *RET*-fp cohort.

Table 2. Biomarkers and molecular characteristics

Co-occurring biomarkers and molecular characteristics*	*RET*-fp (N=26)	Matched *RET*-WT (N=104)	Non-matched *RET*-WT (N=7,116)
TMB status, n (%)			
High (≥20 mut/Mb)	2 (7.7)	0	215 (3.0)
Medium (<20, ≥5.7 mut/Mb)	5 (19.2)	16 (15.4)	1,323 (18.6)
Low (<5.7 mut/Mb)	17 (65.4)	88 (84.6)	5,578 (78.4)
Missing	2 (7.7)	0	0
MSI-high, n (%)			
Yes	1 (3.8)	0	119 (1.7)
No	13 (50.0)	86 (82.7)	6,021 (84.6)
Unknown / Missing	12 (46.1)	18 (17.3)	976 (13.7)
Oncogenic alteration, n (%)			
ALK	0	0	13 (0.2)
BRAF	0	6 (5.8)	389 (5.5)
ERBB2	1 (3.8)	4 (3.8)	300 (4.2)
EGFR	0	0	53 (0.7)
MET	0	0	9 (0.1)
KRAS	0	39 (37.5)	2,622 (36.8)
NTRK	0	0	13 (0.2)
ROS1	0	0	11 (0.2)

*Variants of known/likely functional status. MSI, microsatellite instability; mut/Mb, mutations per megabase; *RET*-fp, *RET* fusion-positive; *RET*-WT, *RET* wild type; TMB, tumour mutational burden.

Survival outcomes

- When the CGP report date was used as the index date (model 1; **Figure 2A**):
 - Median OS was 6.0 months (95% CI 1.6–9.9) in the *RET*-fp cohort and 9.7 months (95% CI 6.3–11.7) in the matched *RET*-WT cohort
 - The hazard ratio (HR) was **1.8** (95% CI 1.1–3.1).
- When the initial diagnosis date was used as the index date (model 2; **Figure 2B**):
 - Median OS was 6.9 months (95% CI 1.6–9.6) in the *RET*-fp cohort and 11.2 months (95% CI 7.7–16.9) in the matched *RET*-WT cohort
 - The HR was **2.2** (95% CI 1.3–3.7).

Figure 2. OS using A) CGP report date as the index date (model 1); B) initial diagnosis date as the index date* (model 2)

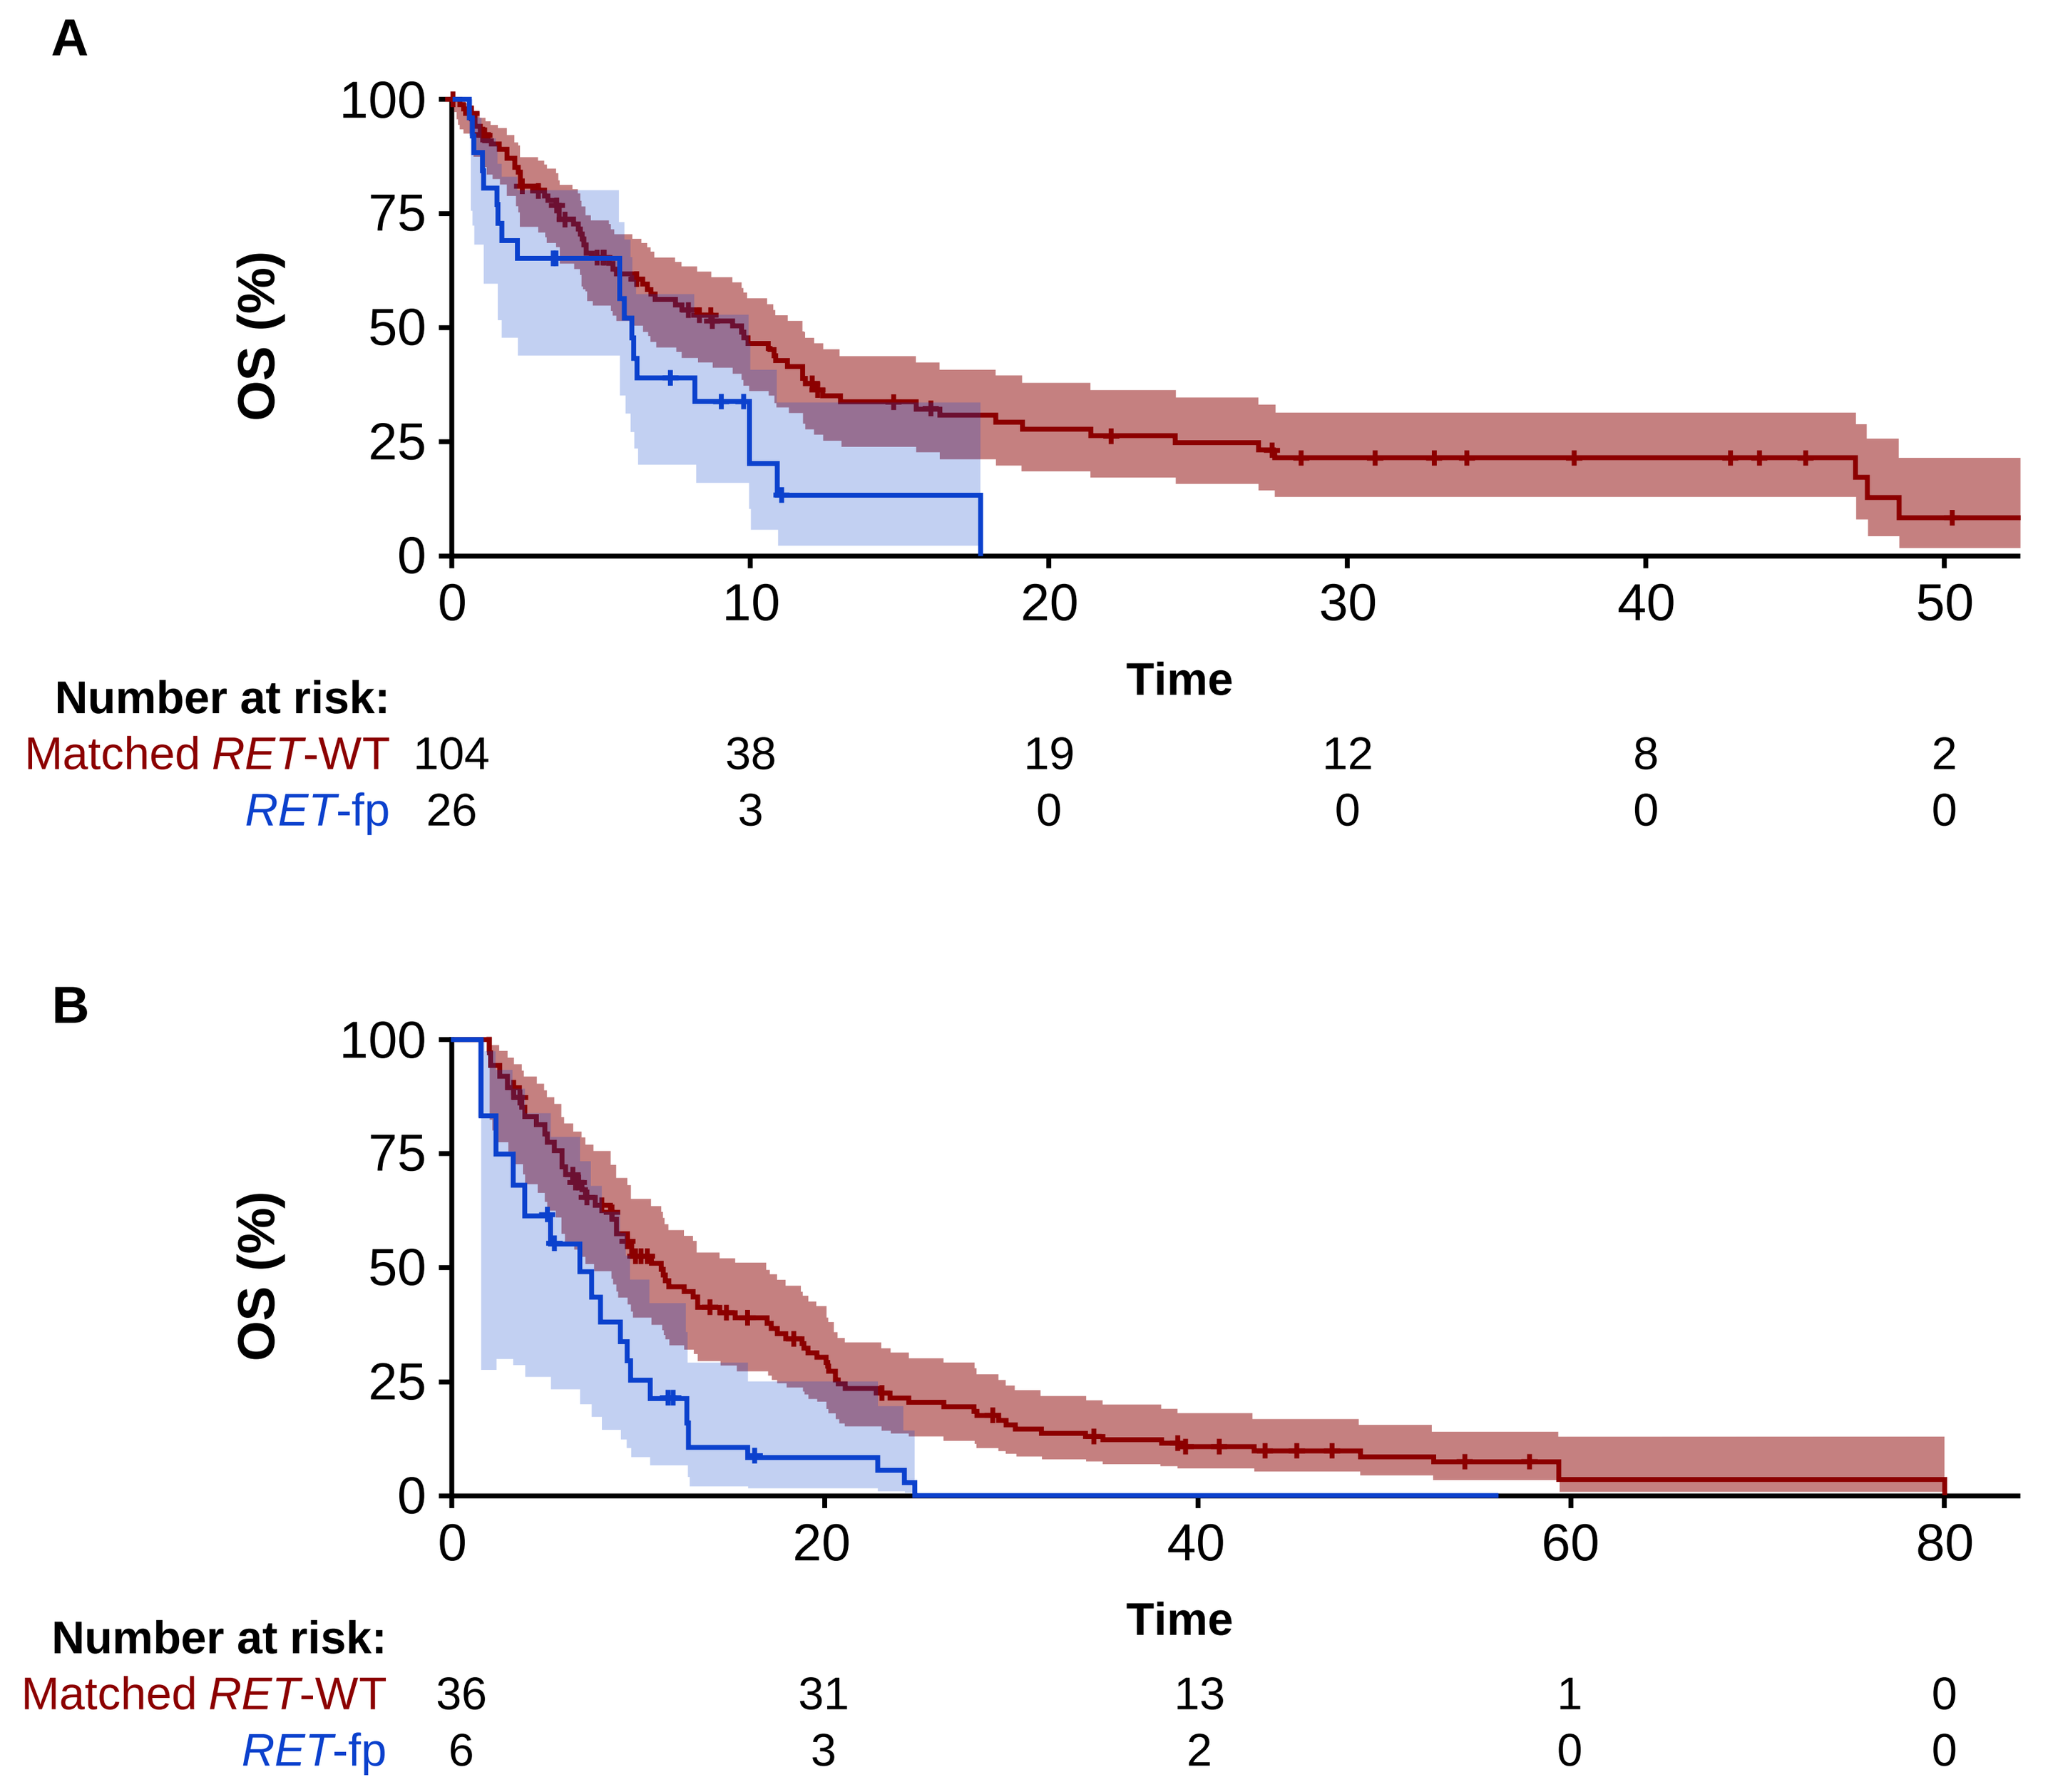
A
OS (%)
100
75
50
25
0
0
10
20
30
40
50
Time
Number at risk:
Matched RET-WT 104 38 19 12 8 2
RET-fp 26 3 0 0 0 0
B
OS (%)
100
75
50
25
0
0
20
40
60
80
Time
Number at risk:
Matched RET-WT 36 31 13 1 0
RET-fp 6 3 2 0 0

*Corrected for immortal time bias.
CGP, comprehensive genomic profiling; OS, overall survival; *RET*-fp, *RET* fusion-positive; *RET*-WT, *RET* wild type.

CONCLUSIONS

- **Despite the small sample size, patients with *RET*-fp solid tumours had a shorter median OS than matched patients with *RET*-WT tumours and may have an increased risk of death.**
- **Oncogenic co-alterations were infrequent in patients with *RET*-fp tumours, which suggests that *RET* fusions are the primary oncogenic drivers in these tumours.**
- **Our data highlight the need for effective RET inhibitors that could improve the survival of patients with solid tumours harbouring *RET* fusions.**

SUMMARY

Median OS: *RET*-fp vs matched *RET*-WT
Model 1: 6.0 vs 9.7 months
Model 2: 6.9 vs 11.2 months

Low TMB and MSI in both the *RET*-fp and matched *RET*-WT cohorts

Low frequency of genetic co-alterations in patients with *RET*-fp solid tumours

Affiliations

[1]Department of Medical Oncology, Ramón y Cajal University Hospital, Madrid, Spain; email address: pilargarridol@gmail.com
[2]Department of Oncology and Hemato-Oncology, Università degli Studi di Milano, and Niguarda Cancer Center, Grande Ospedale Metropolitano Niguarda, Milan, Italy
[3]Earle A. Chiles Research Institute, Providence Portland Medical Center, Portland, Oregon, USA
[4]F. Hoffmann-La Roche Ltd, Basel, Switzerland

Disclosures

Pilar Garrido reports an advisory board role for Abbvie, Amgen, AstraZeneca, Bayer, GlaxoSmithKline, Janssen, Lilly, MSD, Novartis, Pfizer, Roche, Takeda and Sanofi; Speaker role for AstraZeneca, Janssen, MSD, Medscape, Novartis, Pfizer, Roche, Takeda and TouchIme. For co-author disclosures please refer to the abstract: https://oncologypro.esmo.org/

Acknowledgements

Study sponsored by F. Hoffmann-La Roche Ltd. The authors wish to thank Andreas Beringer for his contribution to this poster. Medical writing support for the development of this poster, under the direction of the authors, was provided by Tahmina Alam, MA, of Ashfield MedComms, an Inizio company, and funded by F. Hoffmann-La Roche Ltd.

References

1. Kato, et al. Clin Cancer Res 2017
2. Belli, et al. Clin Cancer Res 2020
3. Drilon, et al. Nat Rev Clin Oncol 2018
4. Singal, et al. JAMA 2019

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without permission of the authors.